Multidimensional Gene Expression Models for Characterizing Response and Metastasis in Solid Tumor Samples

POSTER #126

Authors: Walt Carney, PhD*; Milan Bhagat‡; Natalie A. LaFranzo, PhD†

ABSTRACT

Predicting response to standard of care therapies and new immunotherapies is enabled by tools that quantitatively analyze the complex heterogeneity of the tumor microenvironment. Current methods to do so primarily rely either on single gene or protein markers, and often report solely on one feature of the multifaceted immune cycle. Unlike other molecular approaches, the ImmunoPrism™ Immune Profiling Assay has been built using multidimensional gene expression models validated to accurately identify immune cells in a complex mixture with high correlation to flow cytometry measurements, but accessible for formalin-fixed paraffin-embedded (FFPE) tissues. We will report results showing the assay is highly sensitive, capable of detecting immune cells present to as little as 2% of the specimen, important for critical immune cell types such as M1 and M2 macrophages and Tregs. Further, the assay requires as few as two sections of FFPE solid tumor tissue or as little as 20 ng of total RNA which enables the analysis of rare clinical archives. In this study, we apply the ImmunoPrism assay to a small cohort of patients with non-small cell lung carcinoma (NSCLC) treated with standard of care therapies. Immune profile changes for pre vs. post treatment and primary vs. metastatic tissue are reported in the context of clinical data, such as therapy response. Resulting biomarkers, including a novel machine-learning based multidimensional marker are also reported with predictive accuracy, PPV, NPV, and selection thresholds. In the future, the same technology will be used to investigate cohorts with triple-negative breast cancer (TNBC) and pancreatic adenocarcinoma (ADC), scaling up sample numbers for increased statistical significance.

PATIENT SELECTION

Patient materials for this study were provided by the TriStar Technology Group human tissue repository. Materials were sourced from collaborating centres with the appropriate ethical approvals and banked in a facility operating under ISO-9001 quality systems. For the data presented here, 7 sets of solid tumor tissue from matched pre and post treatment biopsies were analyzed from patients diagnosed and treated for NSCLC.

The samples were all collected within the last 10 years, and represent patients who were treated with standard of care therapies including chemotherapy and radiation.

Patients include both males and females, median age 61 at time of primary diagnosis, all of caucasian descent, with a mixed history of drinking and smoking. For those patients tested, no EGFR or ALK mutations were detected by molecular assays.

Tristar ID	Collection Date	Diagnosis	Age At Diagnosis (years)	Gender	Specimen Type	Cancer status	Treatment	Disease Progression (months)	Survival status
1757-A (Pre)	2011	Adenocarcinoma	83	M	Lung	Primary	Chemo	18	Deceased
1757-B (Post)	2013	Recurrent Adenocarcinoma (Mediastinal)	84		Mediastinum	Recurrence		not stated	
1758-A (Pre)	2012	Adenocarcinoma	65	F	Lung	Primary	Chemo	30	Alive
1758-B (Post)	2014	Metastatic Adenocarcinoma of pulmonary origin	68		Bone	Metastatic		0	
1759-A (Pre)	2013	Adenocarcinoma	50	M	Lung	Primary	Radio	25	Deceased
1759-B (Post)	2015	Recurrent Multifocal Pulmonary Adenocarcinoma	52		Lung	Recurrence		not stated	
1760-A (Pre)	2014	Adenocarcinoma	61	F	Lung	Primary	Radio	28	Alive
1760-B (Post)	2016	Recurrent Lung Adenocarcinoma	63		Lung	Recurrence		18	
1761-A (Pre)	2015	Mucinous Adenocarcinoma	57	M	Lung	Primary	Chemo	16	Alive
1761-B (Post)	2016	Recurrent Moderately differentiated ADCA	59		Lung	Recurrence		not stated	
1762-A (Pre)	2015	Adenocarcinoma	59	F	Lung	Primary	Chemo	16	Alive
1762-B (Post)	2017	Metastatic adenocarcinoma of lung/pulmonary origin	61		Adrenal Gland	Metastatic	Radio	not stated	
1763-A (Pre)	2016	Adenocarcinoma	71	F	Lung	Primary	Chemo	9	Deceased
1763-B (Post)	2017	Recurrent Adenocarcinoma of lung	72		Lung	Recurrence		5	

Table 1. Clinical meta data for individual patients/samples included in this study.

METHODS - ImmunoPrism™ Assay Development and Validation

Database of Health Expression Models

Using machine-learning, Cofactor has developed multidimensional gene expression models of key immune cells known to be important in therapy response.

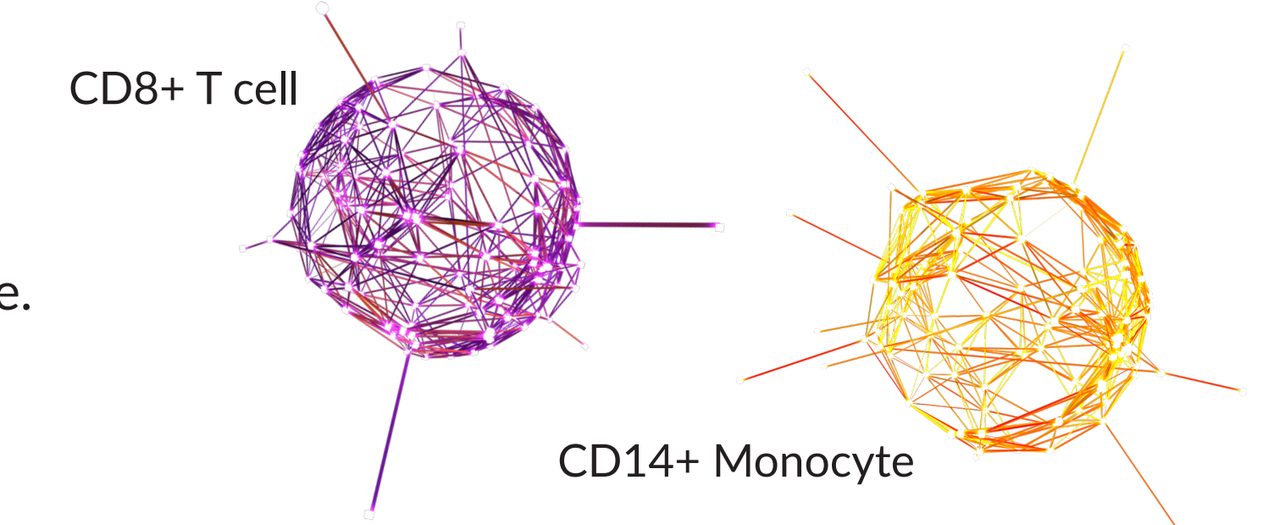
CD8+ T cell
CD14+ Monocyte

Materials and Methods

RNA Extraction and Quality Control: Unstained, unmounted FFPE sections from the same FFPE block were processed for RNA extraction using the Cofactor Prism Extraction Kit, following the manufacturer's suggested protocol. Total RNA was evaluated for quality and quantity using the Bioanalyzer or TapeStation assay (Agilent, Santa Clara, CA), and the Qubit RNA HS or BR Assay (ThermoFisher, Waltham, MA). RNA concentration and quantity (in ng/µL and total ng) and quality (DV200, % of fragments above 200 bp) was evaluated to determine library input amount.

ImmunoPrism Library Preparation and Sequencing: Total RNA was processed for library construction by Cofactor Genomics (http://cofactorgenomics.com, St. Louis, MO) according to the ImmunoPrism Assay standard protocol for FFPE materials. Libraries were sequenced as single-end 75 base pair reads on a NextSeq500 (Illumina, San Diego, CA) following the manufacturer's protocols.

ImmunoPrism Analysis: An individual ImmunoPrism report including expression characterization and immune cell quantification was provided for each sample processed. Samples were grouped according to sponsor-provided clinical meta and outcomes data to generate ImmunoPrism Biomarker Reports, as appropriate.

Analytes Included in Assay: Immune Health Expression Models for CD4+ T cells, CD8+ T cells, CD56+ Natural Killer cells, CD19+ B cells, CD14+ monocytes, Tregs, M1 and M2 macrophages plus the key escape genes PD-1, PD-L1, CTLA4, OX40, TIM-3, BTLA, ICOS, CD47, IDO1, ARG1

Superior Immune Profiling using Health Expression Models

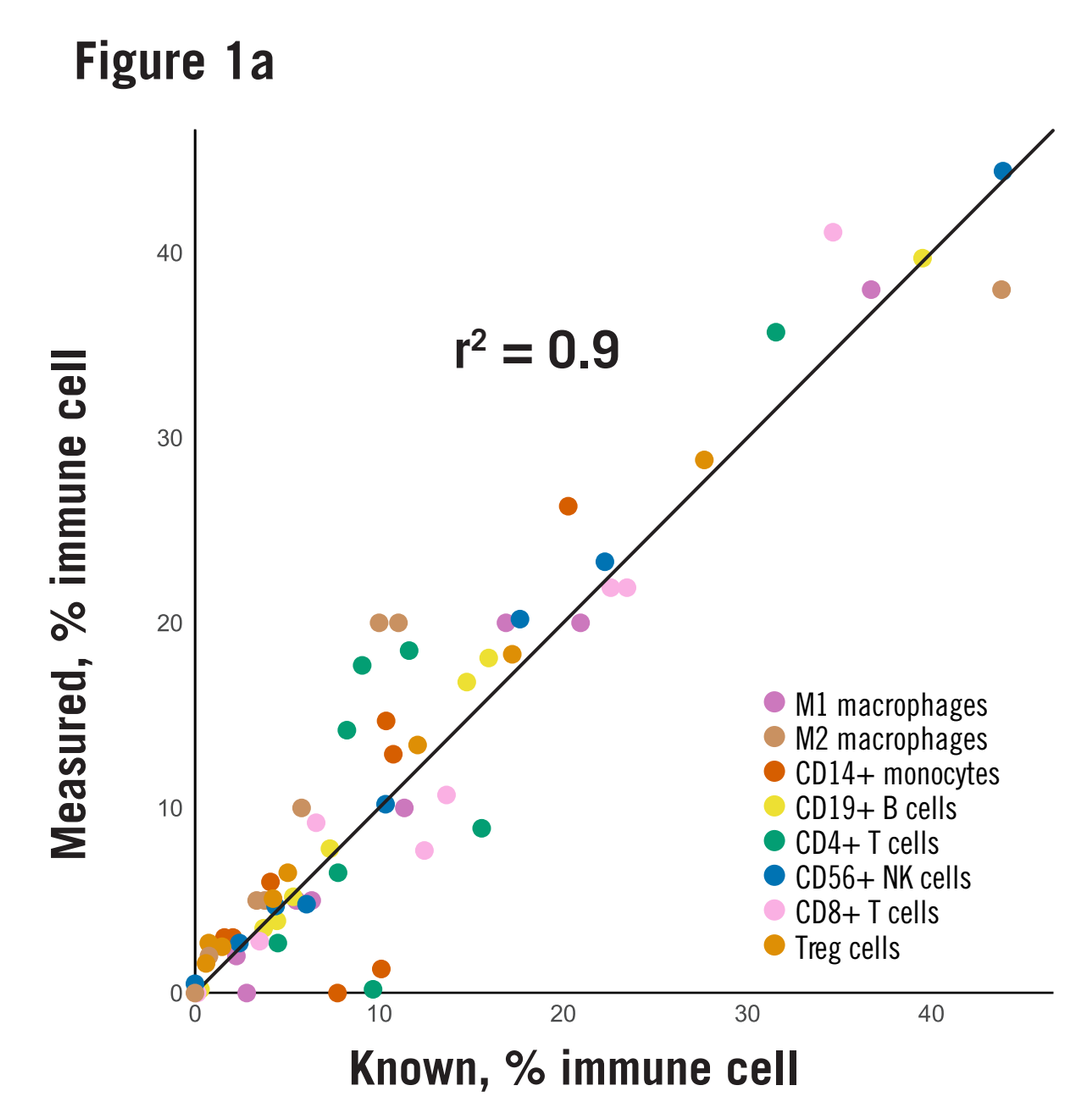
Figure 1a
r² = 0.9
Measured, % immune cell
Known, % immune cell
M1 macrophages
M2 macrophages
CD14+ monocytes
CD19+ B cells
CD4+ T cells
CD56+ NK cells
CD8+ T cells
Treg cells

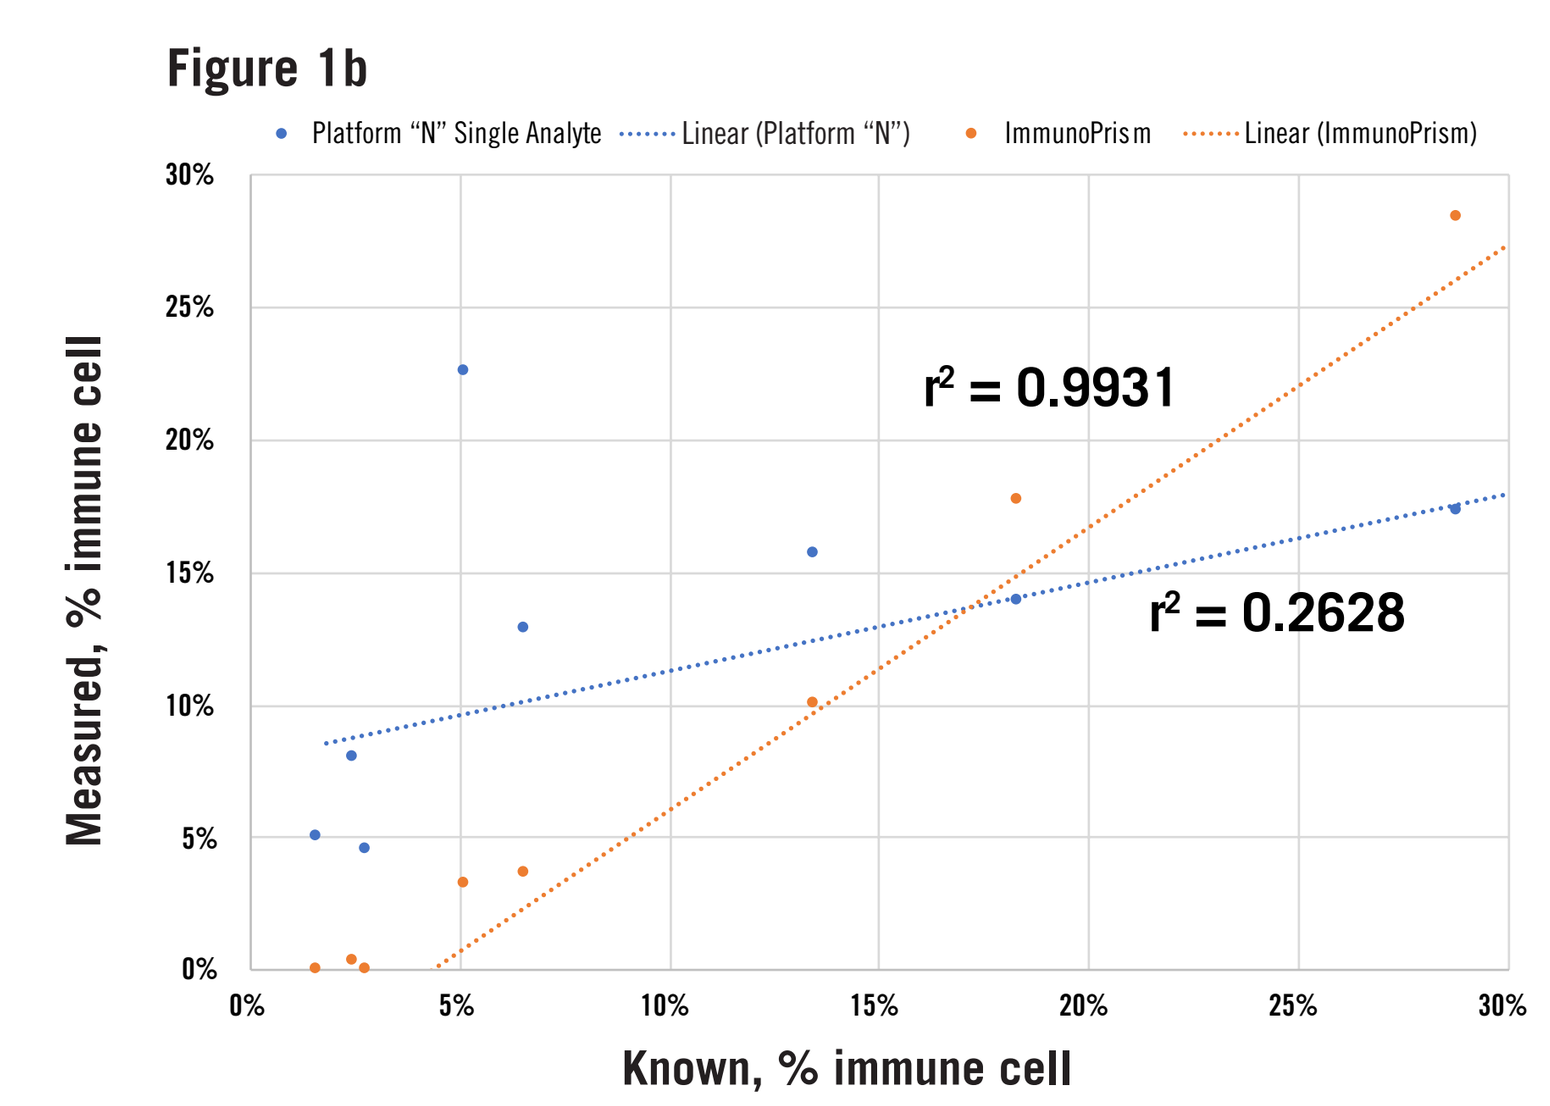
Figure 1b
Platform "N" Single Analyte
Linear (Platform "N")
ImmunoPrism
Linear (ImmunoPrism)
r² = 0.9931
r² = 0.2628
Measured, % immune cell
Known, % immune cell

Using RNA-based Health Expression Models, Cofactor's ImmunoPrism assay is able to quanitfy immune cells in complex mixtures, comparable to flow cytometry (Figure 1a) and with improved performance over single-analyte approaches (Figure 1b) with control cell mixtures.

RESULTS - Predictive Immune Modeling

Tristar ID	Total RNA Quantity, ng	DV_{200}, %
1757-A (Pre)	1680	57
1757-B (Post)	105	27
1758-A (Pre)	1479	62
1758-B (Post)	1116	65
1759-A (Pre)	1860	69
1759-B (Post)	500	67
1760-A (Pre)	1380	66
1760-B (Post)	1158	73
1761-A (Pre)	1086	63
1761-B (Post)	1095	69
1762-A (Pre)	1251	73
1762-B (Post)	1398	73
1763-A (Pre)	602	79
1763-B (Post)	1770	69

Table 2. Molecular QC results for total RNA extracted from two FFPE sections of solid tumor tissue. Quantity and quality are excellent for clinical archives.

	Alive	Deceased
Median Total Immune - Pre Treatment	65%	61%
Median Total Immune - Post Treatment	54%	64%
Median Delta Total Immune - Pre/Post Treatment	-10%	4%

Table 3. Survival Status is correlated to changes in immune content pre and post treatment. For this cohort, the change in total immune content was correlated with the survival outcome, even though the pre-treatment samples did not have a clear immune cell signal.

Note: Delta = median of pre minus post values.
Positive values indicate that the analyte went up post treatment;
Negative values indicate the analyte went down post treatment.

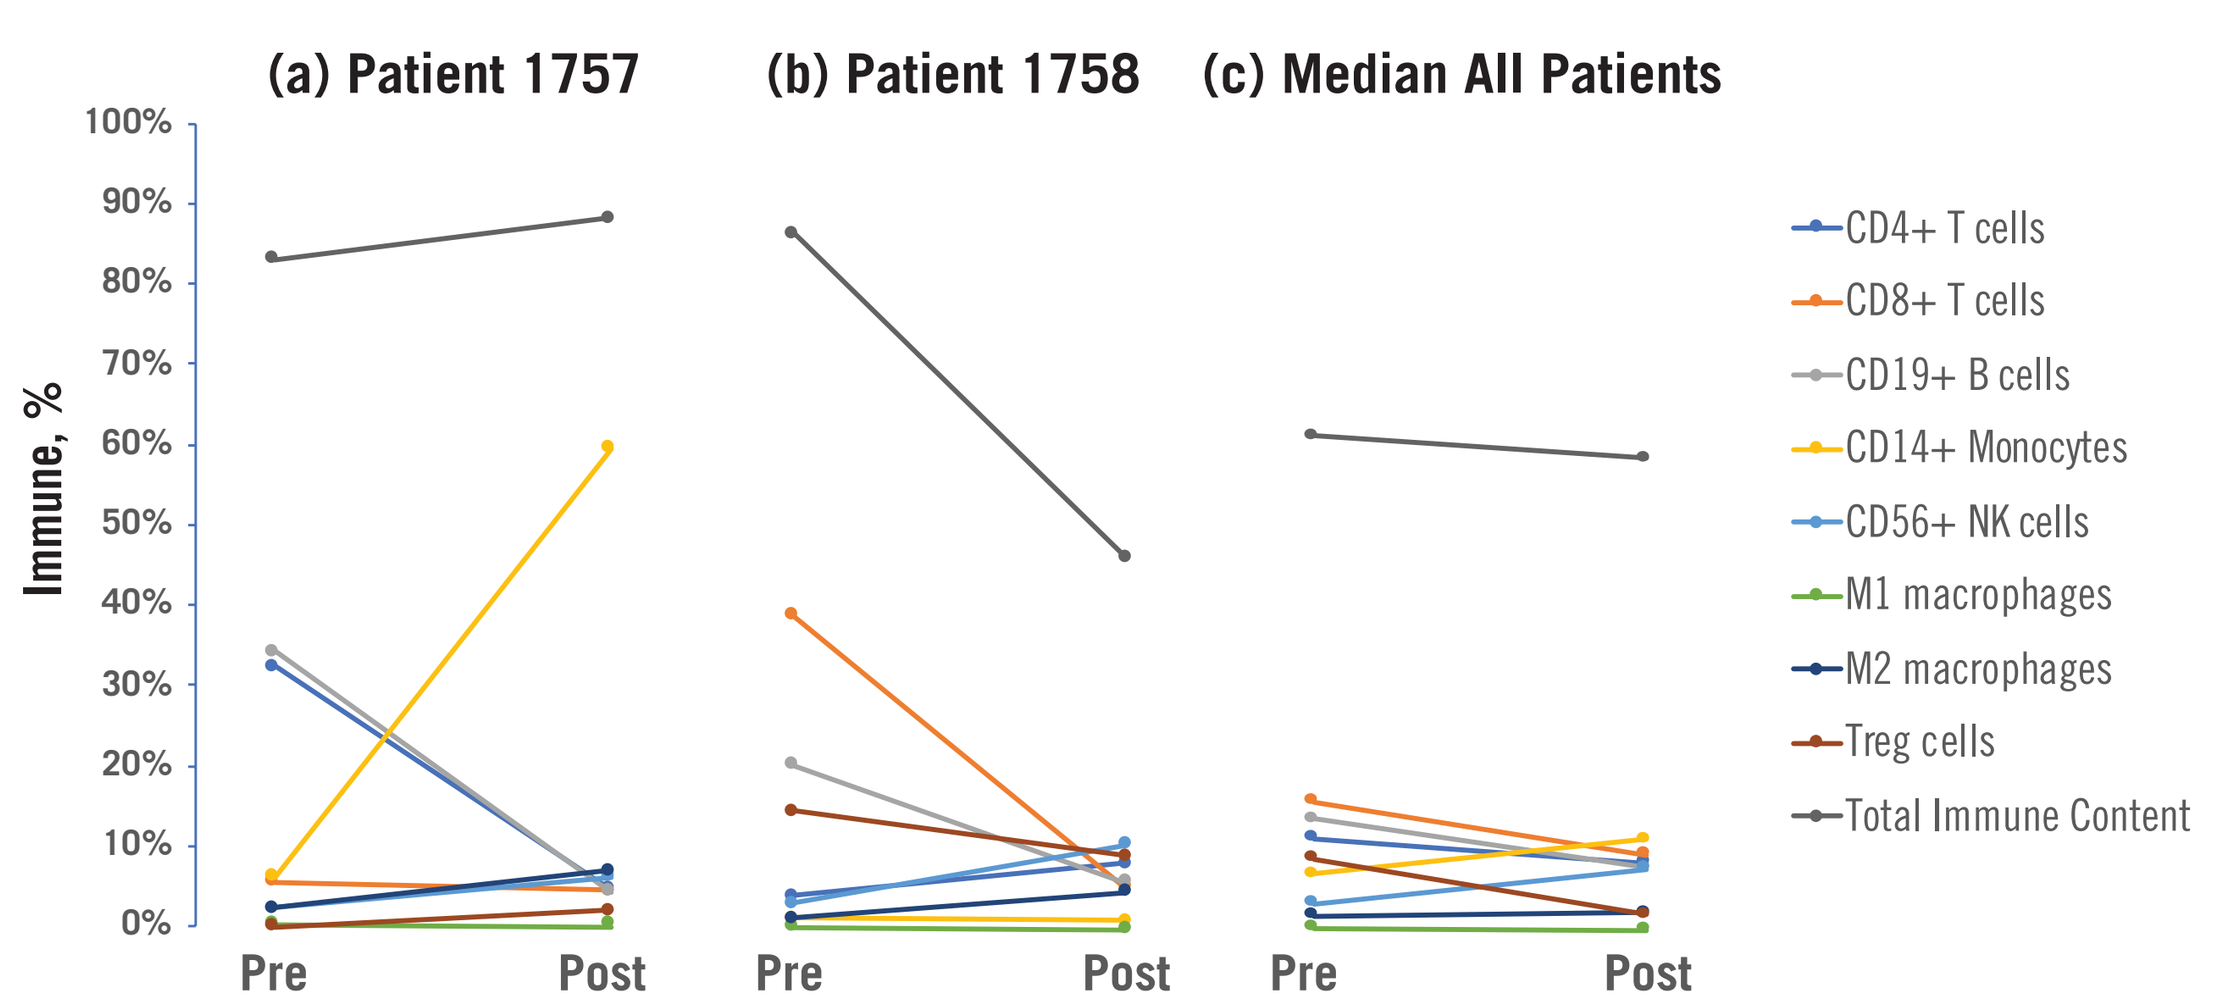
(a) Patient 1757
(b) Patient 1758
(c) Median All Patients
Immune, %
Pre
Post
CD4+ T cells
CD8+ T cells
CD19+ B cells
CD14+ Monocytes
CD56+ NK cells
M1 macrophages
M2 macrophages
Treg cells
Total Immune Content

Figure 2. Individual patient immune profiles from FFPE solid tumor characterization pre and post treatment. Patient 1757 (Figure 2a) showed a significant increase in CD14+ monocytes and decrease in both CD19+ B cells and CD4+ T cells. Patient 1758 (Figure 2b) showed a significant overall decrease in total immune content. On average (Figure 2c) the patients in this study showed slight decreases in immune cells present, however, when factors such as survival status and disease progression are considered, more clear correlations are visible, as shown in the remaining figures.

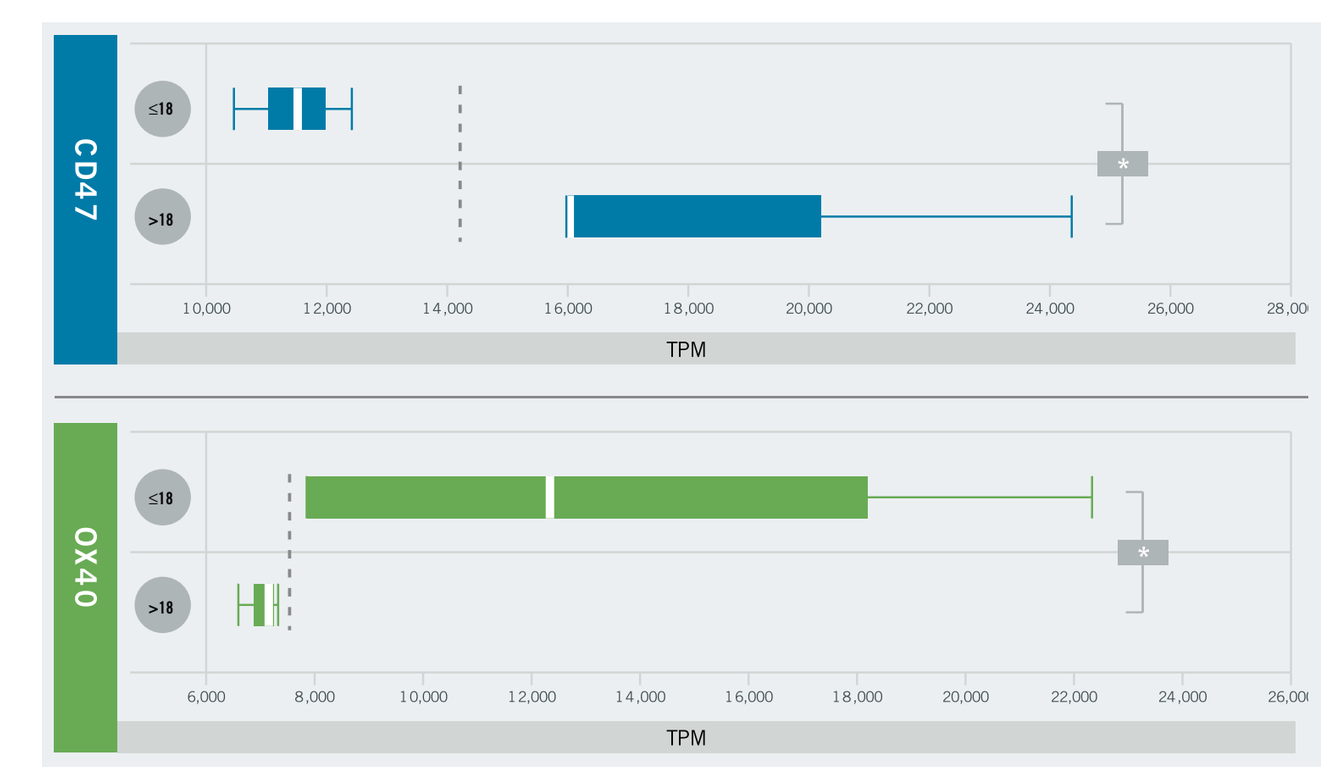
CD47
OX40
≤18
>18
TPM

Figure 3. Disease Progression Biomarkers. In this study, two escape genes, CD47 and OX40, show statistically-significant (p <0.05) differences in the pre-treatment samples for patients whose disease progressed in ≤18 months and those whose disease progressed in >18 months.

Escape genes are quantified using TPM (transcripts per million), a standard unit for gene expression measurement. The left and right sides of the box indicate the 1st and 3rd quartiles of the respective data set. The median is indicated by the white line inside the box. The minimum and maximum inlier datapoints are denoted by the ends of the whiskers. The optimal threshold for predicting outcomes for future samples is shown as a vertical dotted line. Wilcoxon rank sum testing is used to test the null hypothesis that the two groups are sampled from the same distr ibution. Significance of rejecting this hypothesis is denoted with a p value of <0.05, shown as one star. This data is auto-generated by the Prism Analysis software and summarized in the ImmunoPrism Biomarker Report.

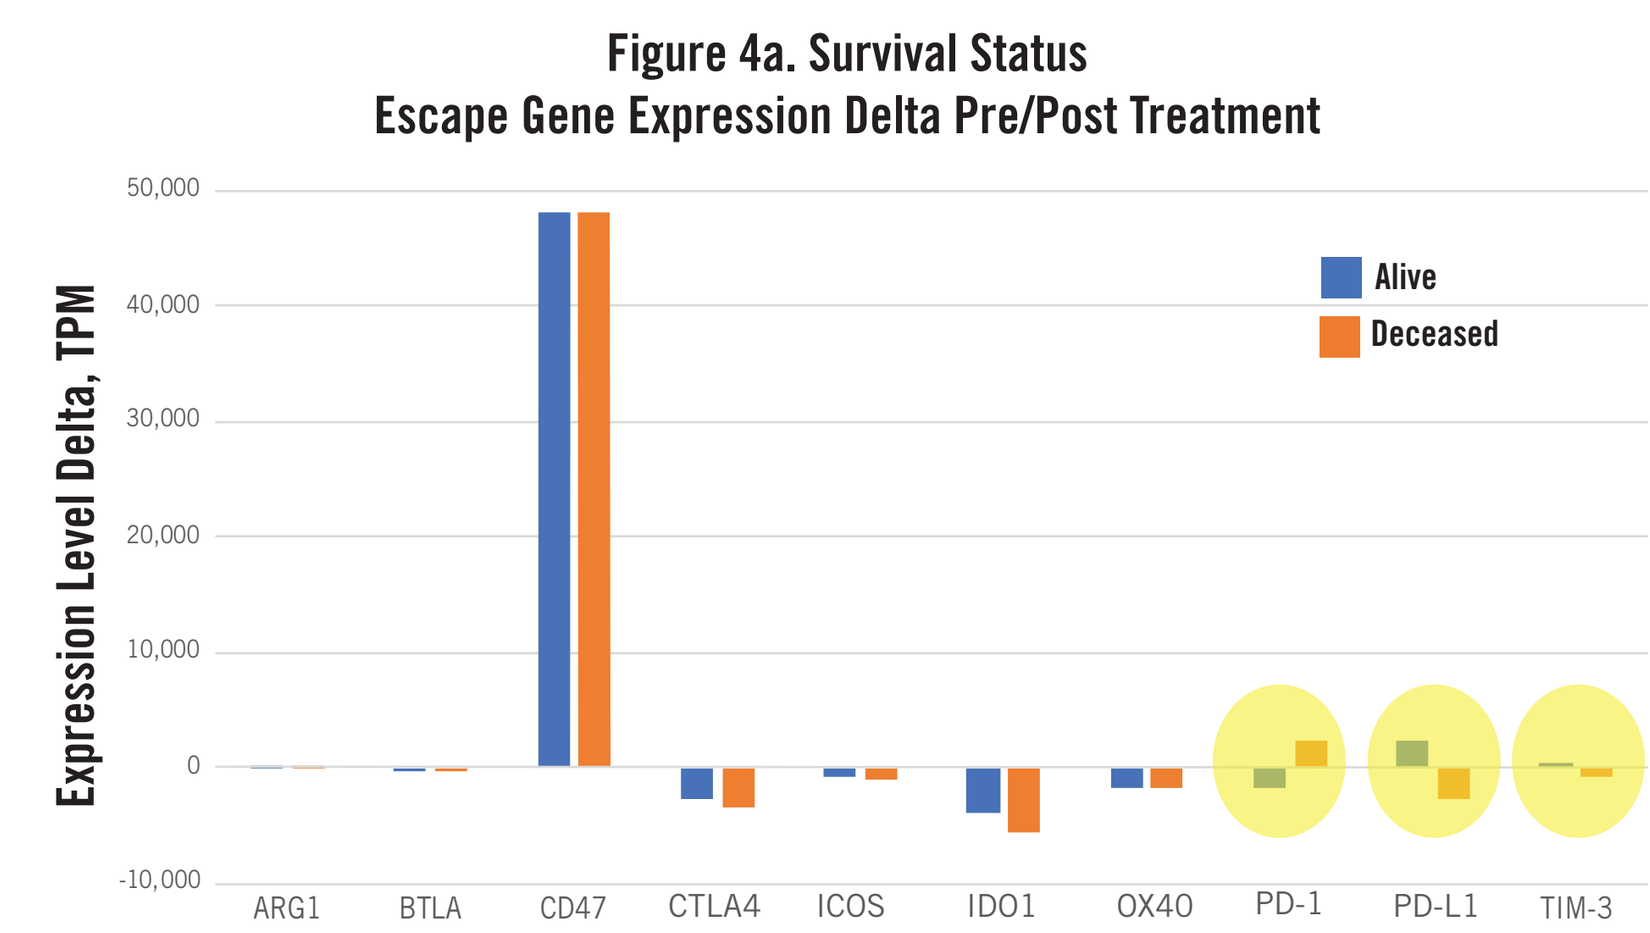
Figure 4a. Survival Status
Escape Gene Expression Delta Pre/Post Treatment
Expression Level Delta, TPM
Alive
Deceased
ARG1
BTLA
CD47
CTLA4
ICOS
IDO1
OX40
PD-1
PD-L1
TIM-3

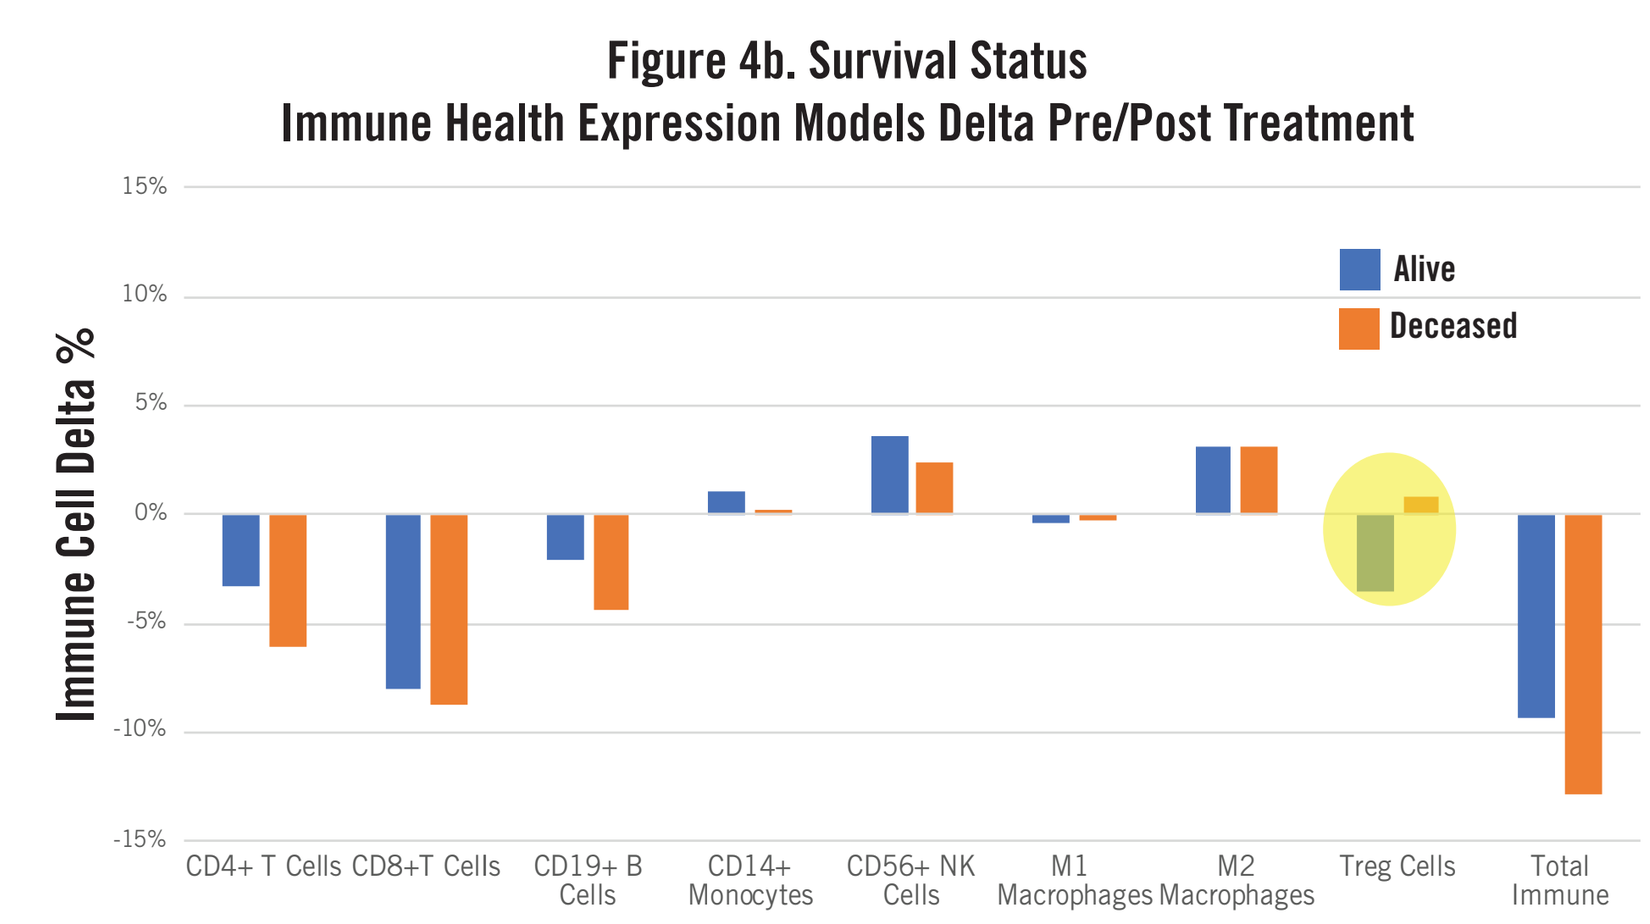
Figure 4b. Survival Status
Immune Health Expression Models Delta Pre/Post Treatment
Immune Cell Delta %
Alive
Deceased
CD4+ T Cells
CD8+T Cells
CD19+ B Cells
CD14+ Monocytes
CD56+ NK Cells
M1 Macrophages
M2 Macrophages
Treg Cells
Total Immune

Figure 4. Survival Status Biomarkers. Quantitative changes between pre and post treatment samples were measured and reported as the "delta" value. Figure 4a summarizes results for escape gene expression and Figure 4b summarizes results for immune cell content. Those highlighted in yellow show clear signal changes between the survival status **(Blue = Alive, Orange = Deceased).**

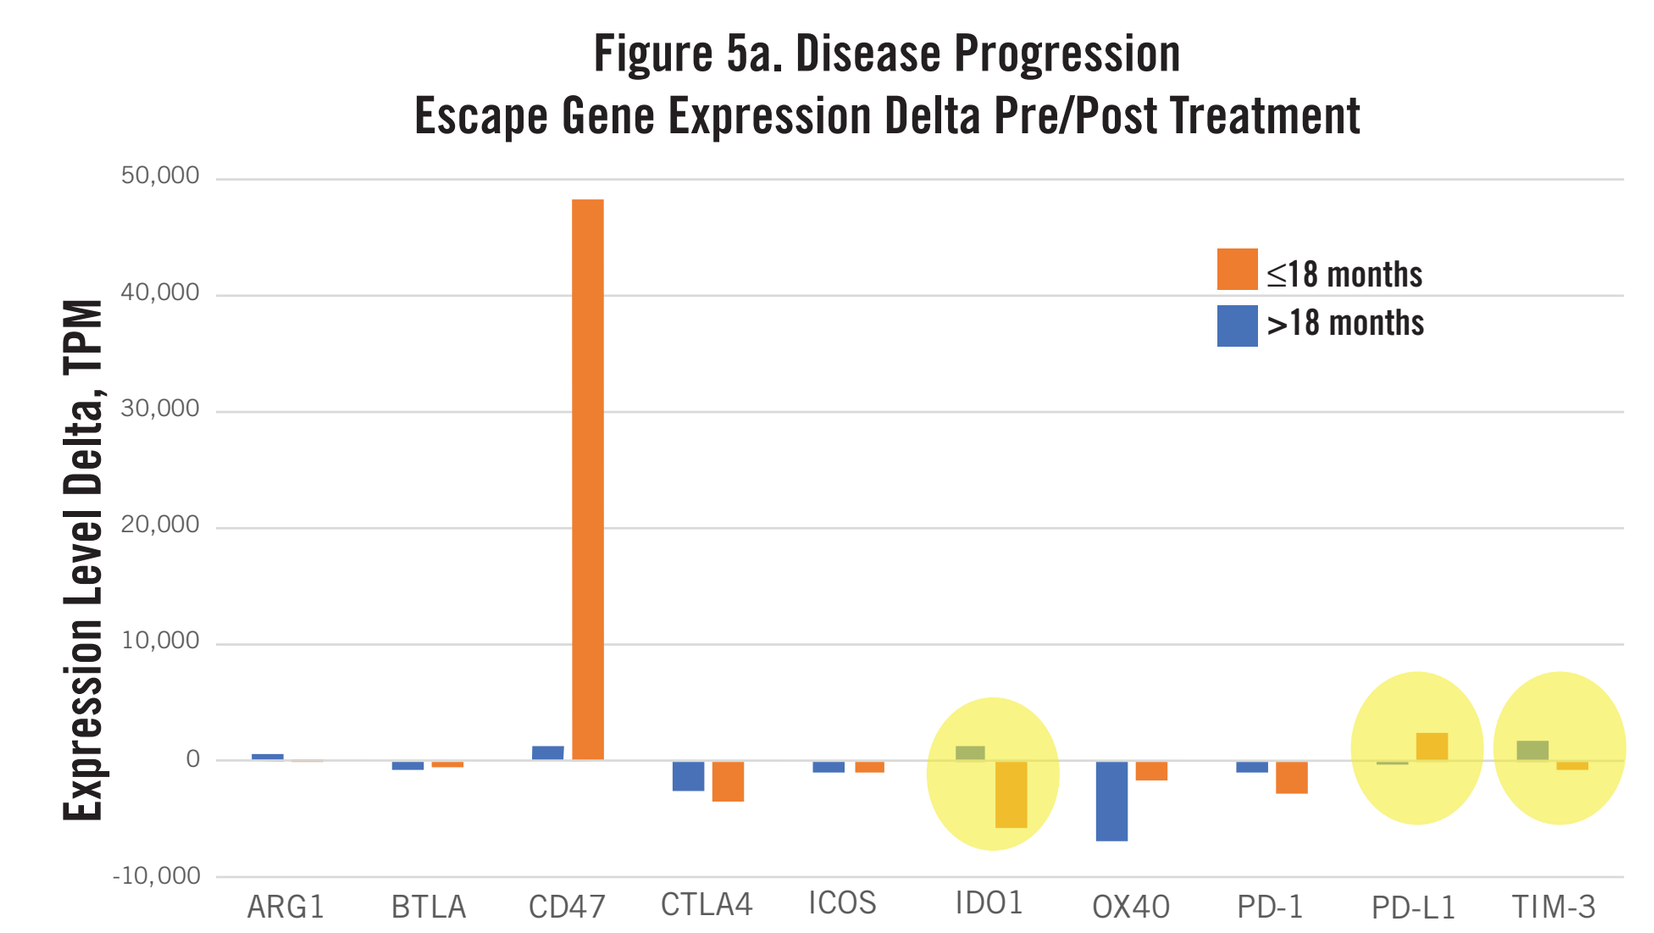
Figure 5a. Disease Progression
Escape Gene Expression Delta Pre/Post Treatment
Expression Level Delta, TPM
≤18 months
>18 months
ARG1
BTLA
CD47
CTLA4
ICOS
IDO1
OX40
PD-1
PD-L1
TIM-3

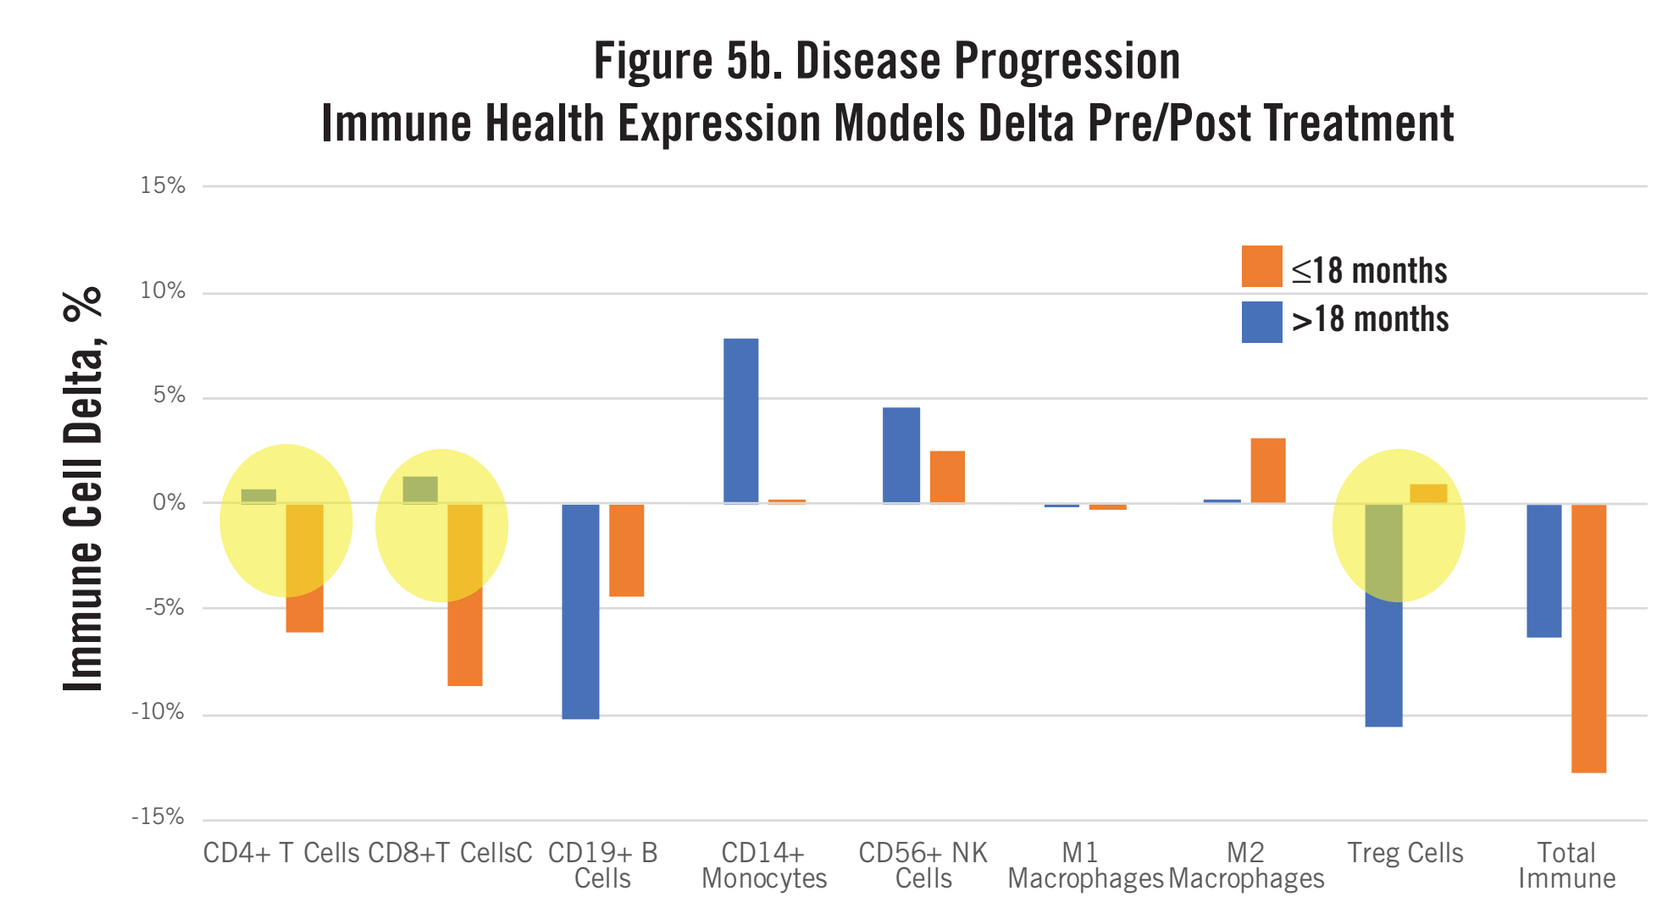
Figure 5b. Disease Progression
Immune Health Expression Models Delta Pre/Post Treatment
Immune Cell Delta, %
≤18 months
>18 months
CD4+ T Cells
CD8+T CellsC
CD19+ B Cells
CD14+ Monocytes
CD56+ NK Cells
M1 Macrophages
M2 Macrophages
Treg Cells
Total Immune

Figure 5. Disease Progression Biomarkers. Quantitative changes between pre and post treatment samples were measured and reported as the "delta" value. Figure 5a summarizes results for escape gene expression and Figure 5b summarizes results for immune cell content. Those highlighted in yellow show clear signal changes between the survival status **(Blue ≤18 months until disease progression, Orange >18 months until disease progression)**.

CONCLUSIONS

Cofactor Genomics' ImmunoPrism assay provides highly quantitative and sensitive measures of immune composition for FFPE solid tumor materials. Requiring only 20 ng total RNA input, the assay maximizes the amount of information accessible from a single FFPE sample, and delivers absolute immune cell quantification which may be compared within and between patients.

TriStar's archived human bio specimens include highly-characterized and well-curated donor samples with extensive clinical follow-up data which enables biomarker discovery and improved disease characterization.

Changes in the immune composition that occur after treatment can be quantified using this method, and may provide insight into immune response to therapy.

Putative biomarkers of disease progression and survival status were identified in this proof-of-concept study including:

- Differences in immune escape gene expression in pre-treatment samples
 - CD47 and OX40
- Changes between pre/post treatment immune profiles (delta)
 - Disease progression:
 - IDO1, PD-L1, PD-1 expression
 - Quantity CD4+ T cells, CD8+ T cells, Treg cells
 - Survival Status:
 - PD-L1, PD-1, and TIM-3 expression
 - Quantity of Treg cells
 - Overall immune cell content

Additional samples will be added to increase statistical power and demonstrate utility of these results.

Clinicians may find utility in the ability to monitor the changing immune landscape in individual patients throughout a treatment regimen, to enable improved decision making for treatment modifications based on biomarkers such as the ones described here. This could be expanded into a more non-invasive approach by correlating blood/serum immune profiles to those observed at the site of the tumor.

FUTURE DIRECTIONS

The promising results seen in this proof-of-concept study with FFPE tissue from non-small cell lung cancer (NSCLC) patients has demonstrated that Cofactor's ImmunoPrism assay is well-suited for characterizing immune profiles in the tumor microenvironment. Further, these results demonstrate that NSCLC patients may benefit from multidimensional biomarkers for predicting response. We intend to scale up to demonstrate stastical and clinical significance by accessing additional specimens from TriStar's databank of well-characterized samples, as well as expand into additional cohorts of triple negative breast cancer patients (TNBC) and pancreatic adenocarcinoma (ADC) treated with standard of care therapies, and peripheral blood mononuclear cells (PBMC) from patients treated with immunotherapies.

Affiliations and Acknowledgements

*Walt Carney Biomarkers Consulting, LLC, Boston, MA USA.
‡: TriStar Technology Group, LLC. Washington, DC 20006, USA.
†: Cofactor Genomics, Inc. San Francisco, CA 94107, USA.
Corresponding author: natalie_lafranzo@cofactorgenomics.com

The authors would like to acknowledge Kevin Flanagan, PhD and Tiange (Alex) Cui, PhD from Cofactor Genomics for their contributions in the molecular and informatics processing of these materials. Copies of this poster may be accessed by the QR code at right; Copies obtained through QR code are for personal use and may not be reproduced without written permission of the authors. Cofactor's ImmunoPrism™ Assay is offered both for Research Use Only (not to be used as a diagnostic assay) and as a CAP-validated assay. All stages of the assay were performed in a research environment for these results. Please contact us to discuss which option is right for your application.